1703-P

Semaglutide reduces multi-organ proteomic-based biological age across cohorts

Maria Dermit[1], **Alejandro Aguayo-Orozco[2]**, Sarah Amandine Caroline Voisin[3], Kasper Adelborg[3], Dylan Belmont-Rausch[4], Helen M. Colhoun[5], Ludger J. E. Goeminne[6], Vadim N. Gladyshev[6], Kristoffer Jensen Kolnes[3], Donna Ryan[7], Lotte Bjerre Knudsen[4]

Scan QR code or visit the web site below for: ePoster slides

https://sciencehub.novonordisk.com/ada2026/aguayo-orozco.html

Aim

- To investigate the effect of semaglutide on biological age using mortality-based proteomic aging clocks.

Introduction

- Semaglutide has shown benefits for multiple clinical endpoints, including reductions in all-cause mortality and major adverse cardiovascular (CV) events (MACE), as well as improvements in kidney outcomes, knee osteoarthritis, and heart failure outcomes in adults with overweight or obesity.[1–6]
- In the SELECT trial, adults with overweight or obesity (body mass index ≥27 kg/m²) and CV disease but no history of diabetes were randomized to once-weekly subcutaneous semaglutide 2.4 mg or placebo.[1]
- Goeminne et al. (2025)[7] developed organ aging models trained to predict time to death using Olink® proteomic data from the UK Biobank.
 - Chronological age, mortality, and longitudinal proteome data were used to train organ-specific and conventional aging models. The models predict organ/system-specific disease and reflect physiologic deterioration.
- In this analysis, the effect of semaglutide on biological age in populations with obesity was investigated using mortality-based proteomic aging clocks.

Methods

- Organ-specific proteomic aging clocks from Goeminne et al. (2025)[7] were applied to Olink® Explore HT proteomic data from SELECT patients, capturing the biological age of individual organs alongside composite systemic clocks.
 - The organismal clock integrates proteomic aging signals across all tissues using 2347 proteins.
 - The multi-organ clock summarizes the mean biological age across organ-specific clocks using 569 proteins.
- Organ clocks particularly relevant to the SELECT cardiometabolic population were assessed (heart, kidney, and adipose clocks) in association with relevant outcomes from the SELECT trial, including MACE (death from CV causes, nonfatal myocardial infarction, or nonfatal stroke), hospitalization for heart failure, and all-cause mortality.
- To determine whether the effects of semaglutide on biological age are body weight dependent or independent, a causal mediation analysis was performed with change in body weight as the mediator.
 - Complementary evidence was drawn from the STEP 1 and STEP 2 trials, which further extends findings to populations with overweight or obesity, without and with type 2 diabetes, respectively.
 - Proteomic data from these trials were assessed using the SomaScan® assay (v4.1).
- The treatment effect of semaglutide was assessed for the organismal and multi-organ clocks.
- Changes in organ clocks were also assessed for their impact on the effect of semaglutide on mortality.

Results

- A total of 1676 patients with Olink® Explore HT proteomic data were included in the analysis (SELECT proteomic clocks subset; **Table 1**).
 - By design, this was a case-enriched subset of patients who experienced incident MACE or heart failure events, resulting in higher baseline inflammatory markers and lower estimated glomerular filtration rate compared with the full trial population.[1]

Biological versus chronological age

- Proteomic biological age showed a strong positive correlation with chronological age (**Figure 1**; R=0.61, p<2.2×10^{-16}).

Organ aging and clinical outcomes

- The heart clock showed its strongest associations with hospitalization for heart failure and CV death, followed by MACE composites and all-cause death (**Figure 2**, red).
- The kidney clock showed its largest effect sizes for macroalbuminuria and nephropathy endpoints, with effect sizes notably exceeding those for all-cause death (**Figure 2**, blue), demonstrating that each clock captures organ-specific disease processes rather than generic mortality risk.

Table 1. Baseline characteristics of patients in the SELECT proteomic clocks subset

Characteristic	Semaglutide (n=791)	Placebo (n=885)
Age, years	62.5 ± 8.9	62.9 ± 9.2
Female sex, %	25	25
White race, %	88	88
Body weight, kg	98.9 ± 18.6	98.5 ± 18.5
BMI, kg/m²	33.7 ± 5.3	33.7 ± 5.2
Current/former smoker, %	21	19
SBP, mmHg	131.9 ± 15.9	130.8 ± 15.0
DBP, mmHg	79.9 ± 10.4	78.3 ± 10.4
History of hypertension, %	85	83
CAD, %	83	86
eGFR, mL/min/1.73 m²	80.0 ± 17.9	79.8 ± 18.5
hsCRP, mg/L, median (IQR)	2.0 (1.0–4.7)	1.9 (0.9–4.7)

Data are mean ± standard deviation unless otherwise stated.
BMI, body mass index; CAD, coronary artery disease; DBP, diastolic blood pressure; eGFR, estimated glomerular filtration rate; hsCRP, high-sensitivity C-reactive protein; IQR, interquartile range; SBP, systolic blood pressure.

Semaglutide and organ aging

- In the SELECT proteomic clocks subset (using Olink® data) (**Figure 3**, left panel), semaglutide significantly reduced heart and kidney biological age at week 20 versus placebo. For both organs, the effect was driven by the average direct effect, suggesting direct biological aging deceleration beyond what can be explained by weight loss alone.
- In contrast, the adipose clock showed a significant increase in biological age at week 20 attributable to the average causal mediation effect, consistent with adipose tissue remodeling driven by fat mass reduction rather than intrinsic aging acceleration.
 - Multi-organ clocks showed reductions at this early time point.
- Using SomaScan®, which extends to later time points and includes STEP 1 and STEP 2 (week 68) alongside SELECT (week 104), the multi-organ clock was significantly reduced across all three trials, while the organismal clock showed a consistent but nonsignificant trend in the same direction as the Olink® data. (**Figure 3**, right panel).

Organ aging and the effect of semaglutide on mortality

- Heart clock changes significantly mediated the effect of semaglutide on all-cause death and CV death (red, false discovery rate <0.05), accounting for approximately 36% of the all-cause mortality benefit (**Figure 4**).
- Kidney clock changes did not significantly mediate any outcome, consistent with its primary prognostic specificity for kidney rather than CV endpoints (**Figure 4**).

Figure 1. Biological versus chronological age in SELECT

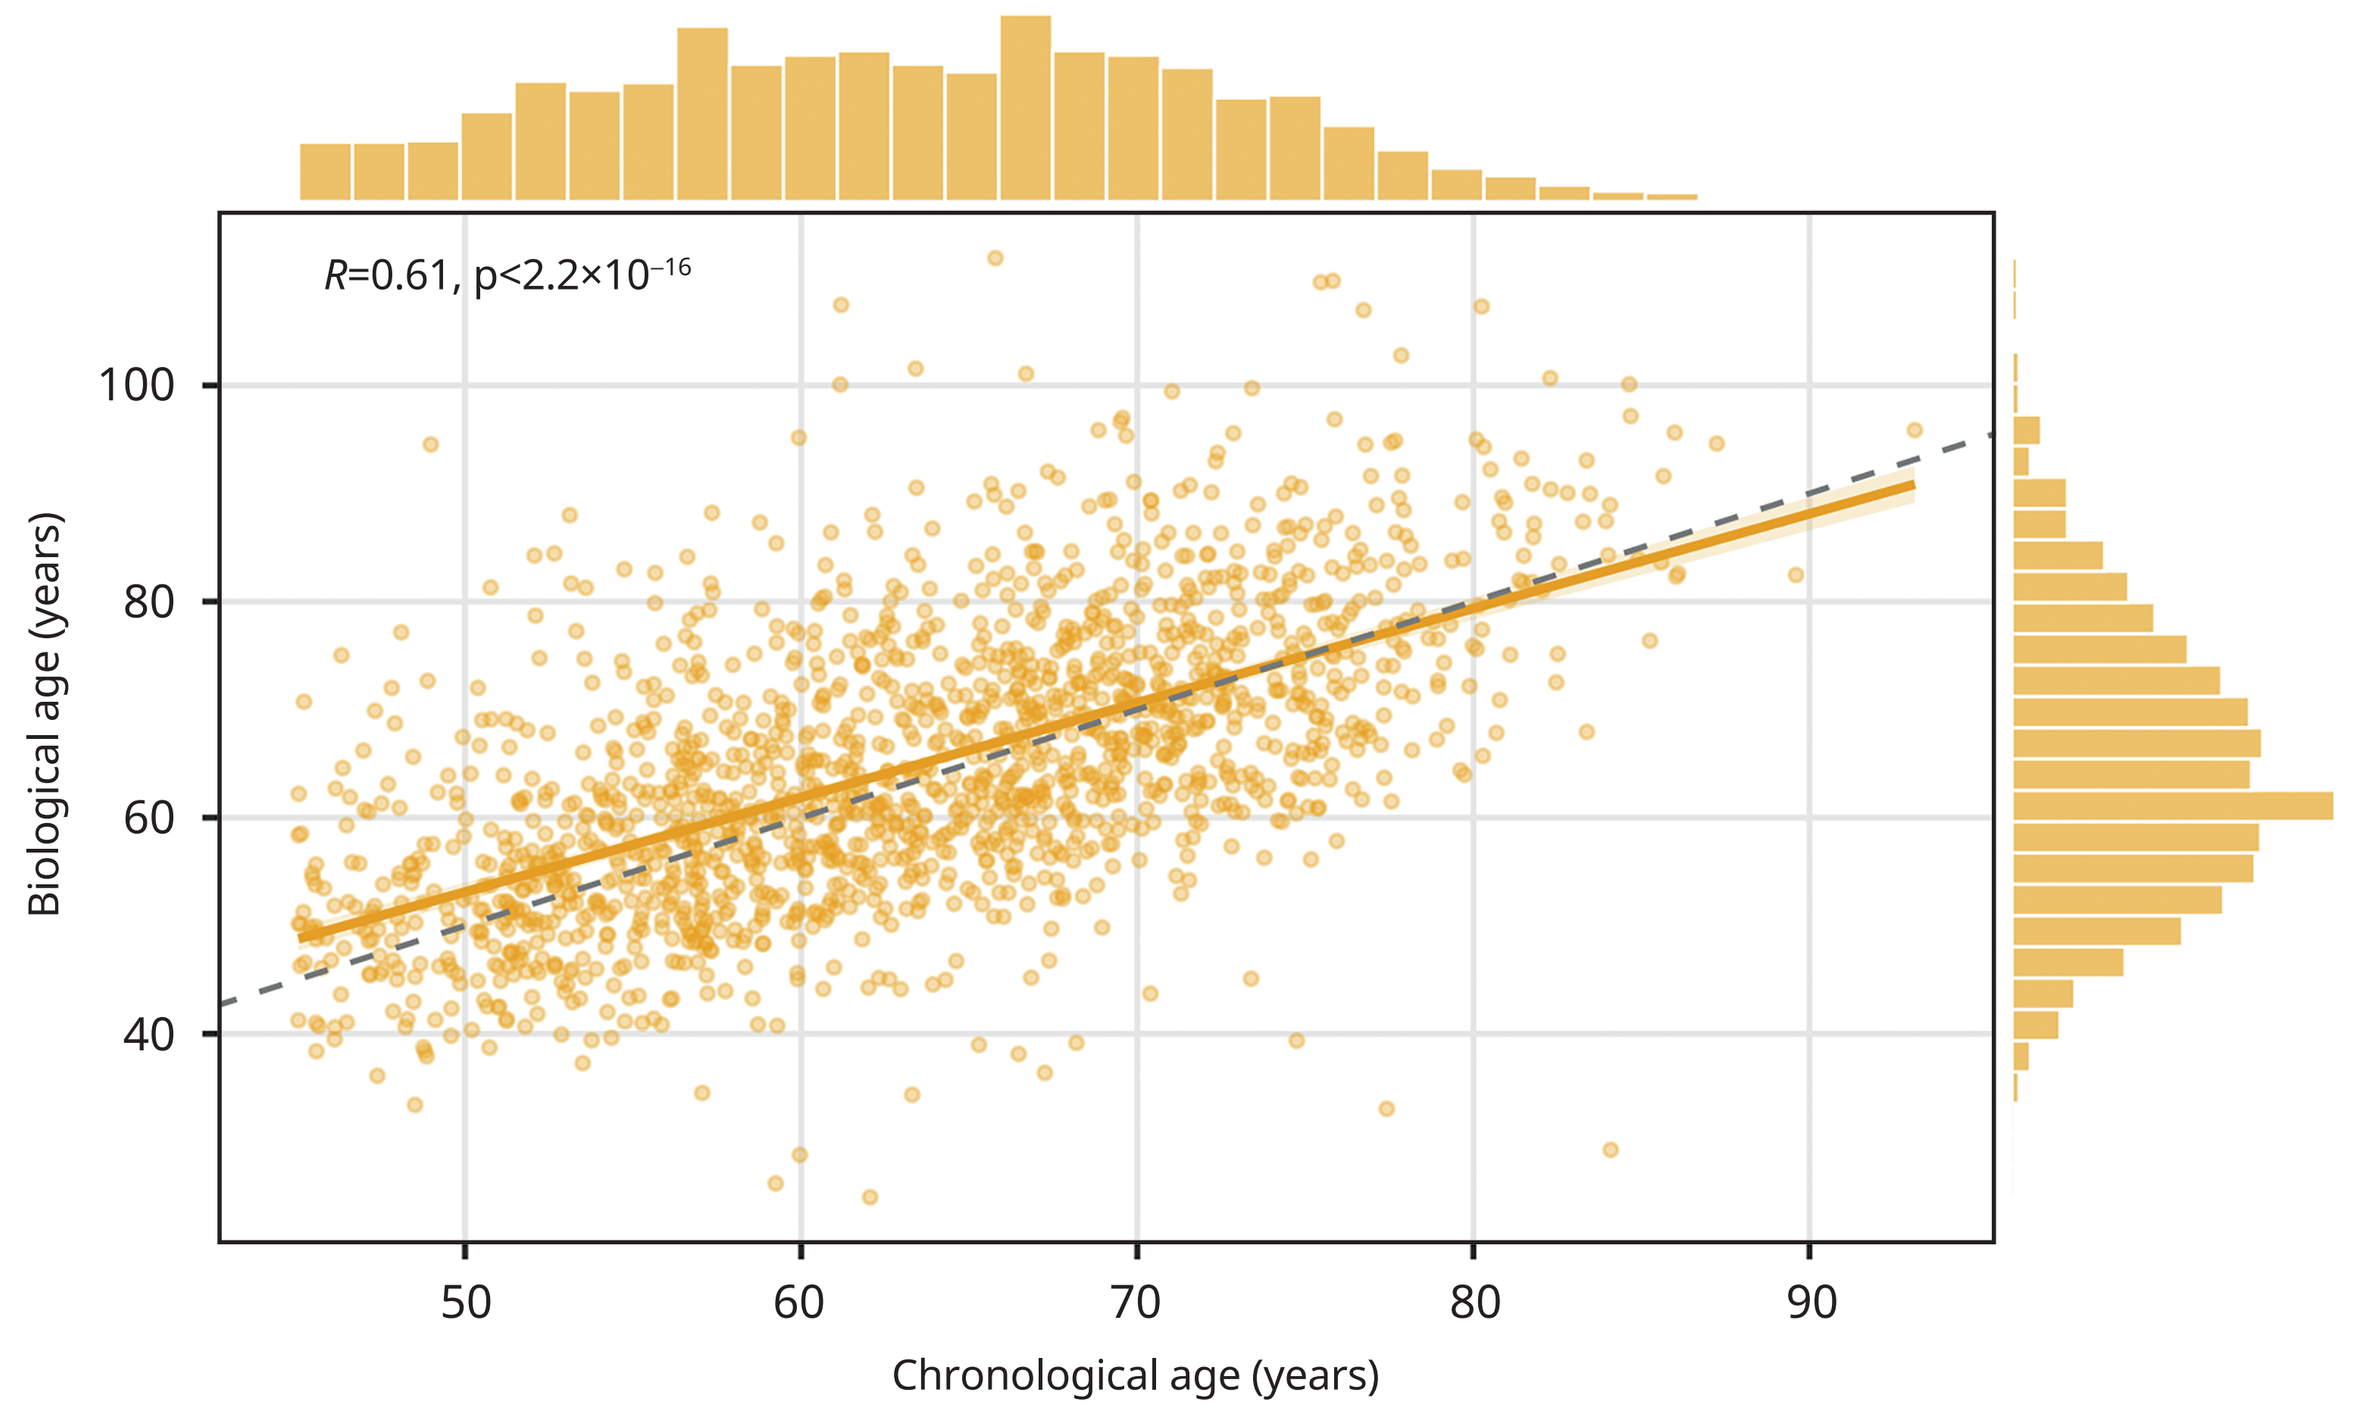
R=0.61, p<2.2×10⁻¹⁶
Biological age (years)
Chronological age (years)
40
60
80
100
50
60
70
80
90

Substantial inter-individual spread around the regression line indicates meaningful divergence between biological (conventional model) and chronological age at every age group, with some patients appearing substantially older or younger biologically than their chronological peers.

Figure 2. Organ aging clocks predict clinical outcomes in SELECT

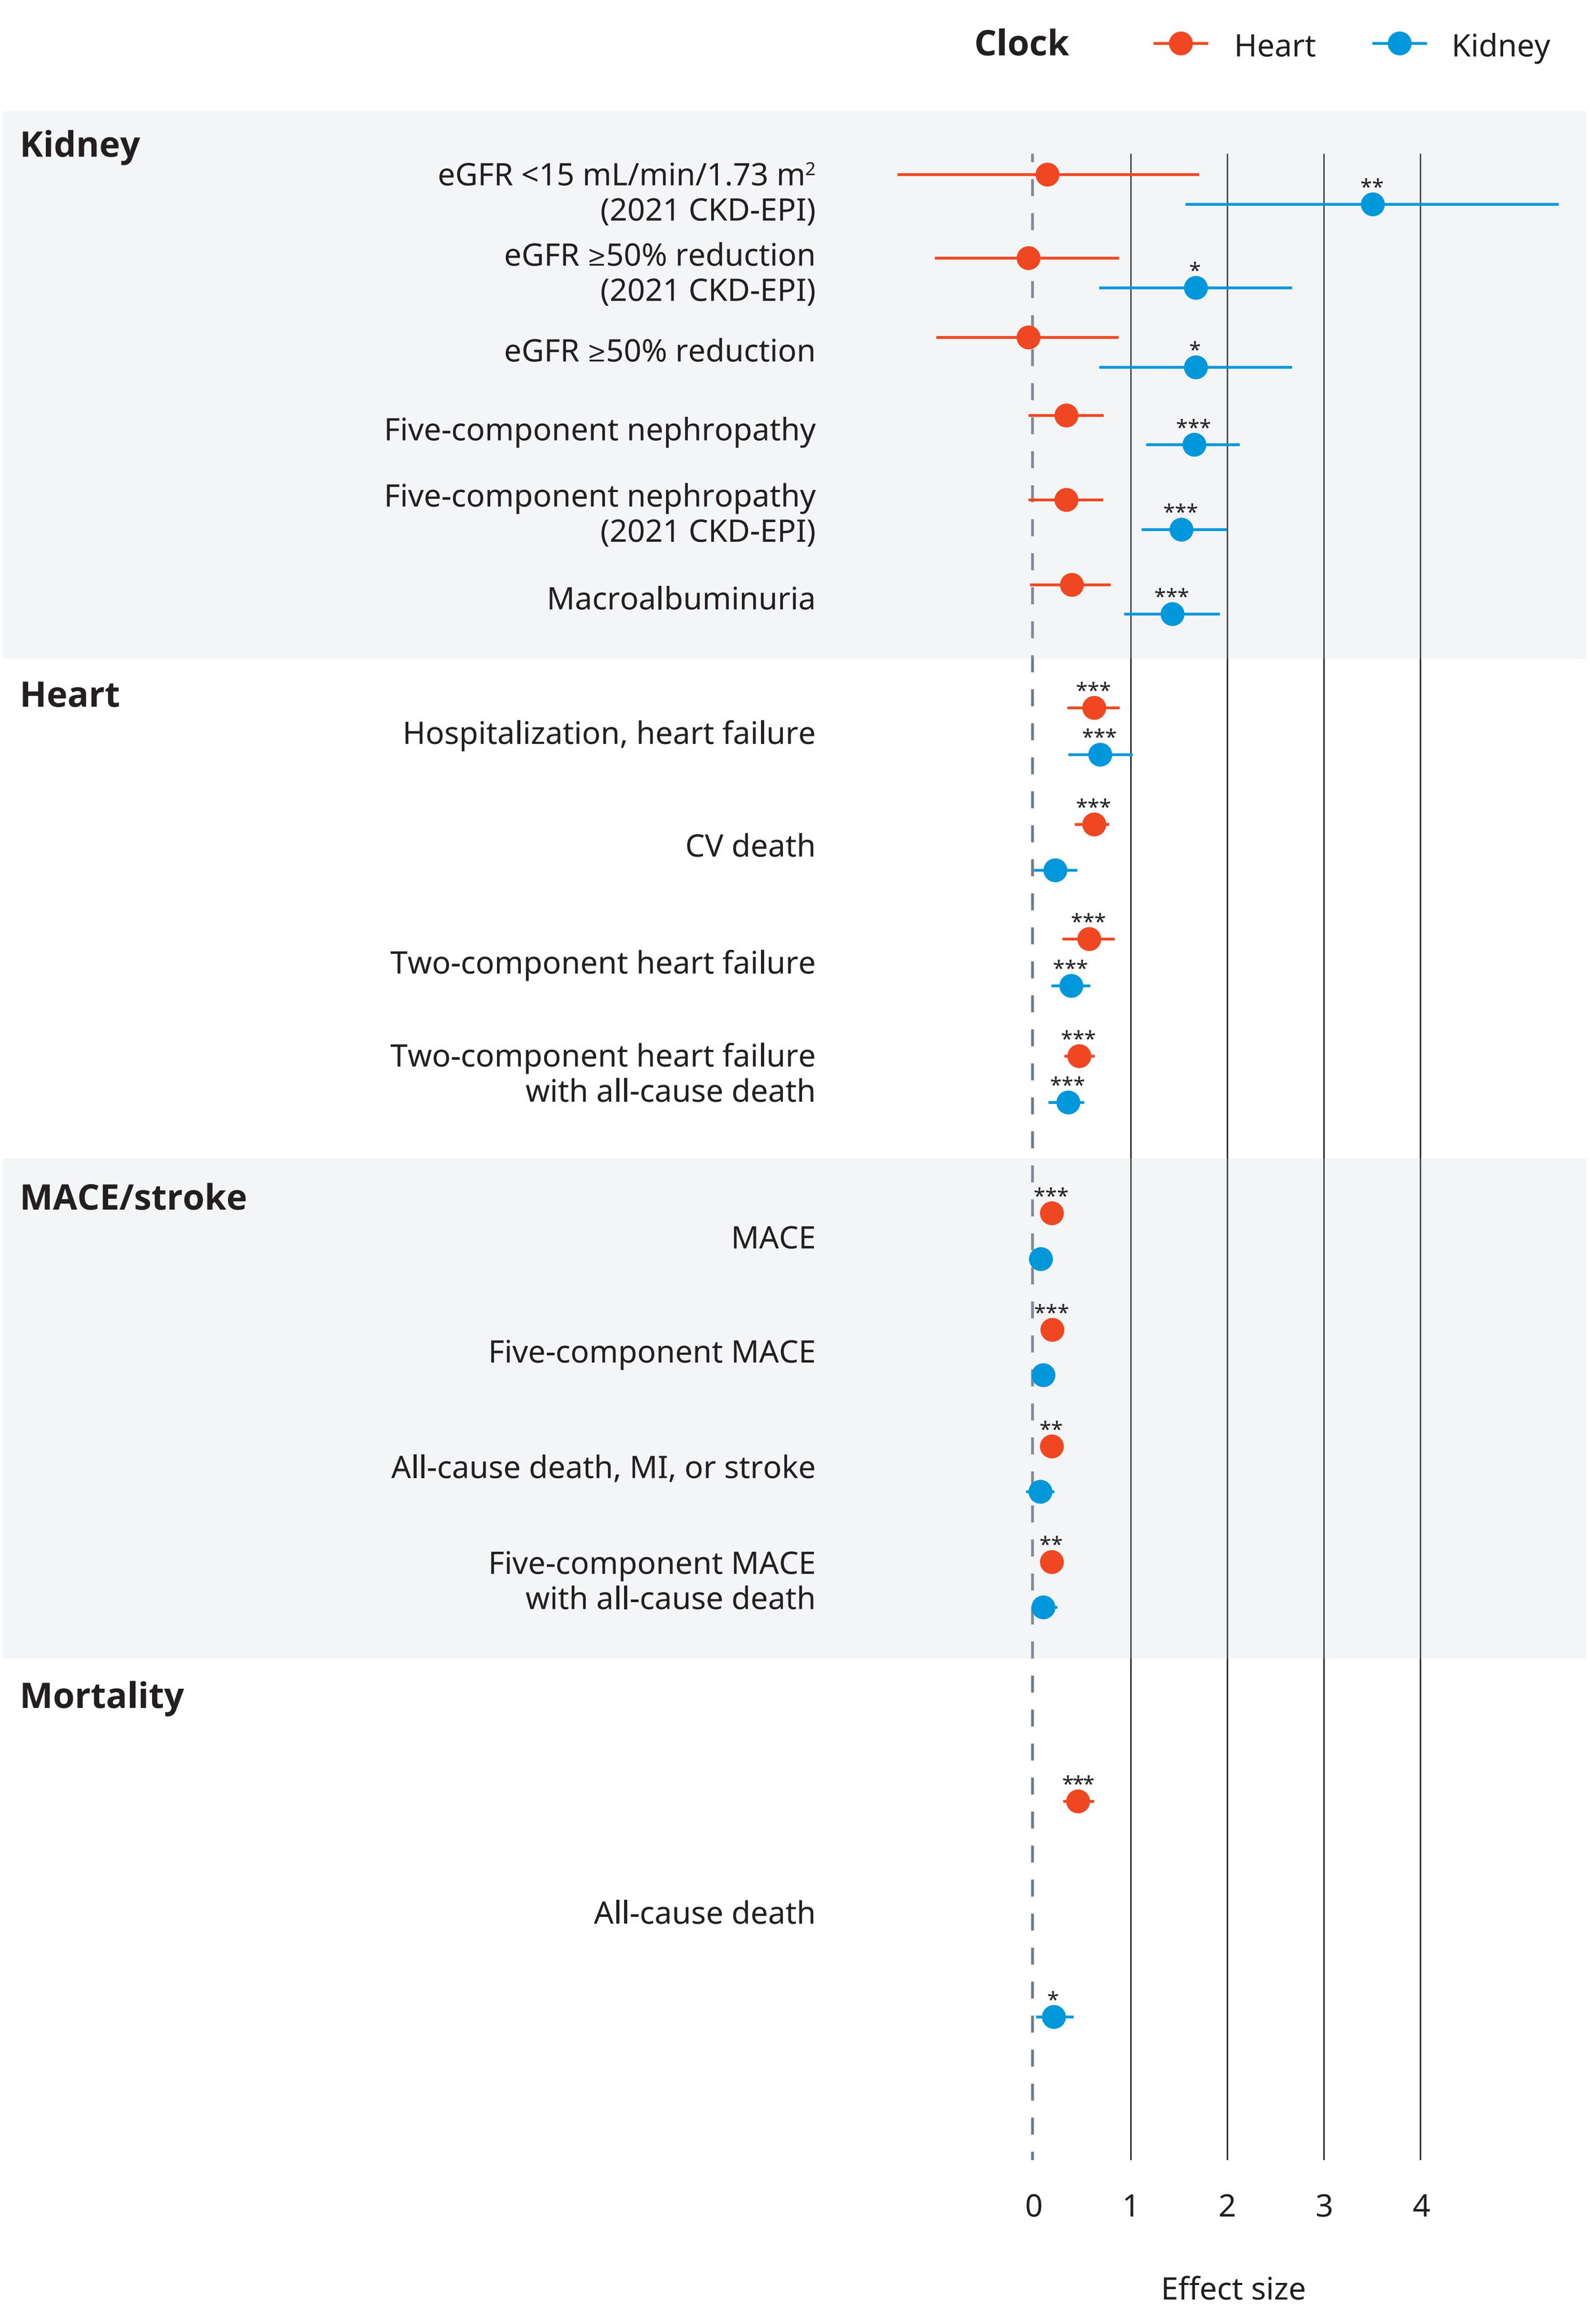
Clock
Heart
Kidney
Kidney
eGFR <15 mL/min/1.73 m² (2021 CKD-EPI)
eGFR ≥50% reduction (2021 CKD-EPI)
eGFR ≥50% reduction
Five-component nephropathy
Five-component nephropathy (2021 CKD-EPI)
Macroalbuminuria
Heart
Hospitalization, heart failure
CV death
Two-component heart failure
Two-component heart failure with all-cause death
MACE/stroke
MACE
Five-component MACE
All-cause death, MI, or stroke
Five-component MACE with all-cause death
Mortality
All-cause death
0
1
2
3
4
Effect size

The heart clock consists of seven proteins (top contributors: MYL4, NPPB, NMRK2, TNNI3, and ITGB1BP2), reflecting myocardial stress, contractile integrity, and metabolic adaptation. The kidney clock consists of nine proteins (top contributors: HAVCR1, REN, UMOD, CYP24A1, and BSND), reflecting tubular injury, renal filtration, and collecting duct function. The adipose clock consists of five proteins (FABP4, LEP, CD300LG, PLIN1, and ADIPOQ), comprising canonical adipokines and lipid-handling proteins reflecting high tissue specificity. Expanded five-component MACE included CV death, nonfatal MI, nonfatal stroke, coronary revascularization, or hospitalization for unstable angina. The heart failure composite included first occurrence of death from CV causes, or hospitalization or an urgent medical visit for heart failure. The main five-component kidney composite endpoint included death from kidney causes, initiation of chronic kidney replacement therapy (dialysis or transplantation), onset of persistent eGFR <15 mL/min/1.73 m², persistent ≥50% reduction in eGFR compared with baseline, or onset of persistent macroalbuminuria.
Significance levels: *p<0.05, **p<0.01, ***p<0.001. Error bars represent 95% CI.
CI, confidence interval; CKD-EPI, Chronic Kidney Disease Epidemiology Collaboration; CV, cardiovascular; eGFR, estimated glomerular filtration rate; MACE, major adverse cardiovascular events; MI, myocardial infarction.

Figure 3. Semaglutide reduces organ biological age in SELECT, STEP 1, and STEP 2

Key result

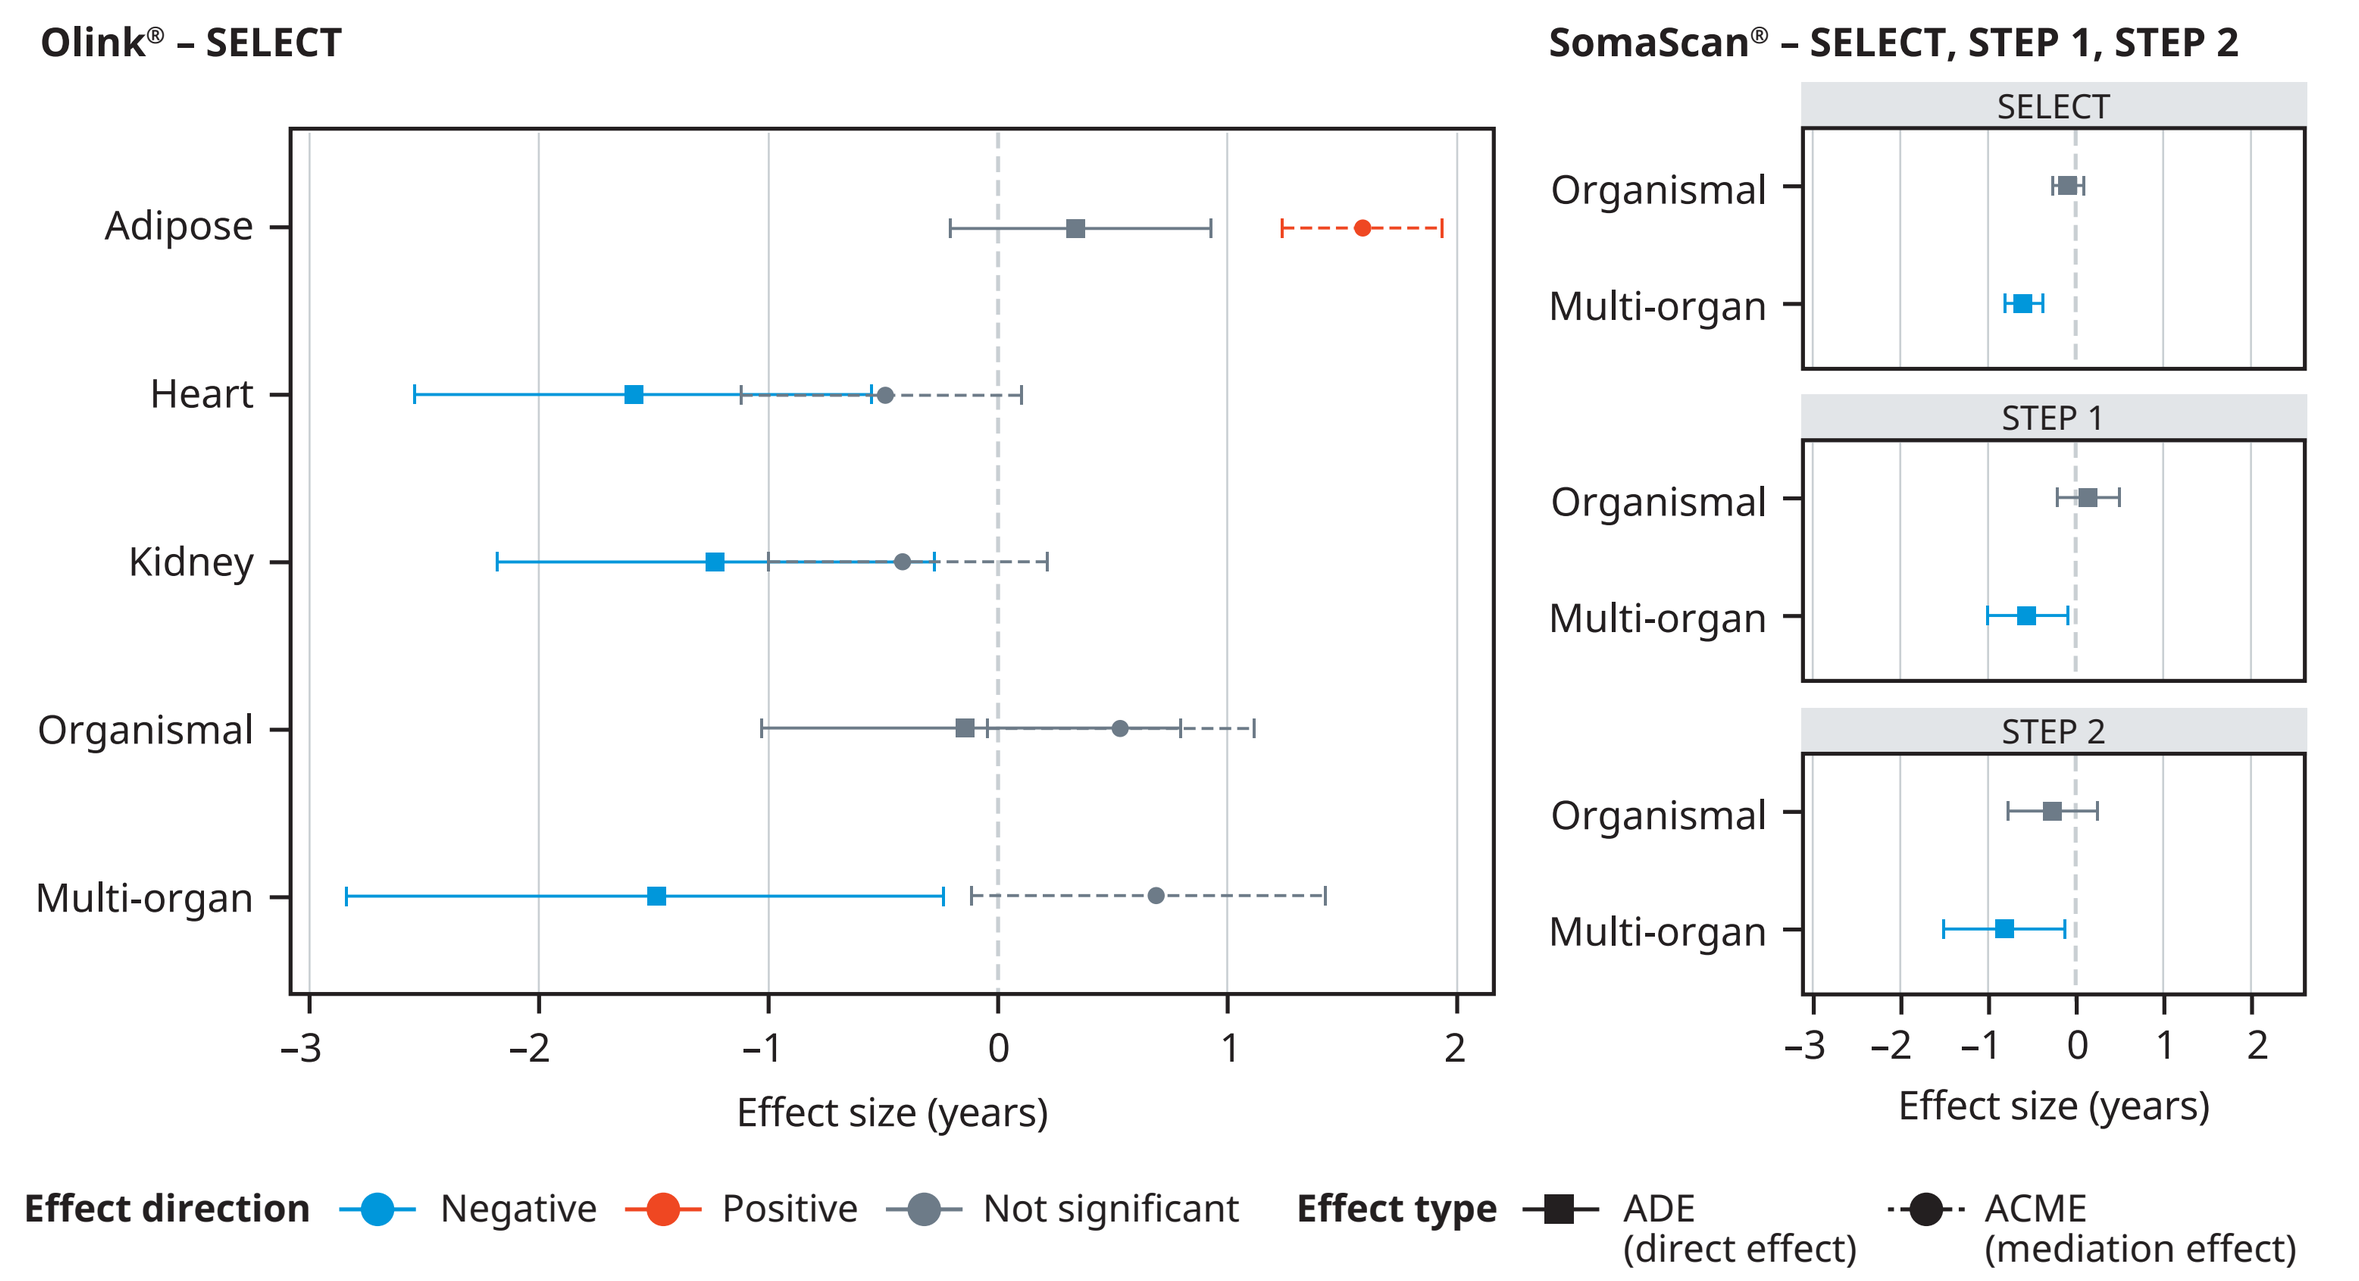
Olink® – SELECT
Adipose
Heart
Kidney
Organismal
Multi-organ
Effect size (years)
SomaScan® – SELECT, STEP 1, STEP 2
SELECT
STEP 1
STEP 2
Organismal
Multi-organ
Effect size (years)
Effect direction
Negative
Positive
Not significant
Effect type
ADE (direct effect)
ACME (mediation effect)

Mediation analysis was performed using the mediation R package. Effect sizes are expressed in biological age years. ACME captures the proportion of the effect of semaglutide attributable to weight loss; ADE reflects the remaining direct effect. STEP 1 and STEP 2 data represent week 68; SELECT SomaScan® data represent week 104, following platform harmonization. Error bars represent 95% CI.
ACME, average causal mediation effect; ADE, average direct effect; CI, confidence interval.

Figure 4. Organ clock changes mediate the effect of semaglutide on mortality

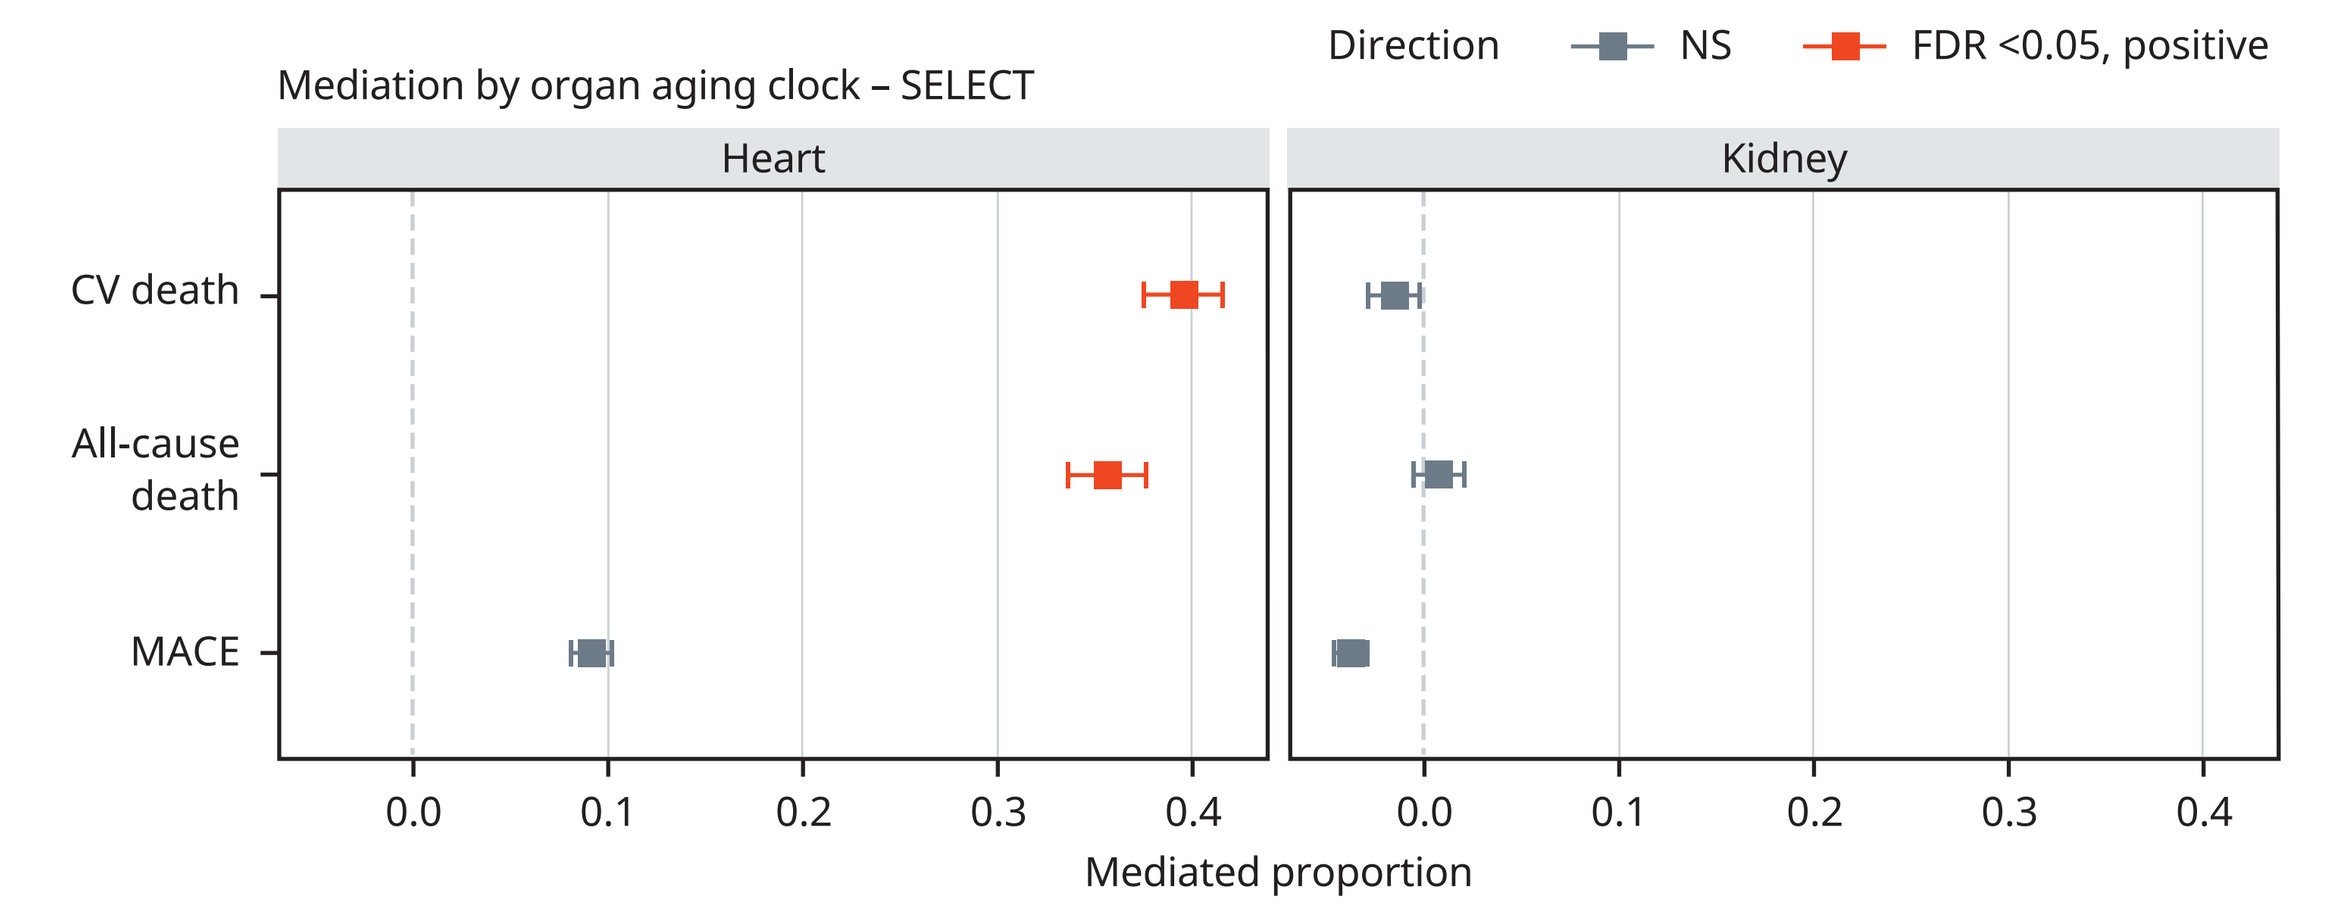
Direction
NS
FDR <0.05, positive
Mediation by organ aging clock – SELECT
Heart
Kidney
CV death
All-cause death
MACE
0.0
0.1
0.2
0.3
0.4
Mediated proportion

Mediation analysis was performed using an Imai/Dai framework. Semaglutide-induced changes in heart and kidney organ aging clock scores at week 20 in SELECT were modeled as mediators of treatment effects on CV and mortality outcomes. The mediated proportion represents the fraction of the total effect of semaglutide on each outcome attributable to clock score change. Significance was assessed using FDR-corrected p<0.05. Error bars represent 95% CI.
CI, confidence interval; CV, cardiovascular; FDR, false discovery rate; MACE, major adverse cardiovascular events; NS, not significant.

Conclusion

- Semaglutide consistently reduced mortality-trained proteomic multi-organ biological age across patients with obesity and different comorbidities, whereas organismal clocks were unchanged. These findings suggest that semaglutide modifies proteomic signatures linked to mortality risk.

[1]Novo Nordisk Research Centre, Oxford, UK; [2]Novo Nordisk A/S, Bagsværd, Denmark; [3]Novo Nordisk A/S, Søborg, Denmark; [4]Novo Nordisk A/S, Måløv, Denmark; [5]Institute of Genetics and Cancer, The University of Edinburgh, Edinburgh, UK; [6]Brigham and Women's Hospital, Harvard Medical School, Boston, MA, USA; [7]Pennington Biomedical Research Center, Baton Rouge, LA, USA.

This analysis was sponsored by Novo Nordisk A/S and all trials are registered with ClinicalTrials.gov (NCT03574597 [SELECT], NCT03548935 [STEP 1], NCT03552757 [STEP 2]). Medical writing support was provided by Roxanna Munir, PhD, of Apollo, OPEN Health Communications, and funded by Novo Nordisk A/S, in accordance with Good Publication Practice (GPP) guidelines (ismpp.org/gpp-2022).
Alejandro Aguayo-Orozco is an employee of Novo Nordisk.
Presented at the American Diabetes Association (ADA) 86th Scientific Sessions, June 5–8, 2026, New Orleans, LA, USA.

References:
(**1**) Lincoff AM et al. *N Engl J Med* 2023;389:2221–2232; (**2**) Colhoun HM et al. *Nat Med* 2024;30:2058–2066; (**3**) Wilding JPH et al. *N Engl J Med* 2021;384:989–1002; (**4**) Davies M et al. *Lancet* 2021;397:971–984; (**5**) Bliddal H et al. *N Engl J Med* 2024;391:1573–1583; (**6**) Kosiborod M et al. *N Engl J Med* 2023;389:1069–1084; (**7**) Goeminne LJE et al. *Cell Metab* 2025;37:205–222.